Safety, Tolerability, Pharmacokinetics (PK), and Pharmacodynamics (PD) of a Dual GLP-1/GIP Receptor Agonist (HRS9531) in Healthy Subjects: A Phase 1, Randomized, Double-blind, Placebo-controlled, Single and Multiple Ascending Dose (SAD and MAD) Study

American Diabetes Association
83RD SCIENTIFIC SESSIONS
SAN DIEGO | JUNE 23–26, 2023
763-P

Kun He[1], Fang Wei[1], Hong Chen[2], Yanyan Zhang[2], Zhen Sheng[2], Qing Wen[1]

[1]Department of Clinical Research Center, Central Hospital Affiliated to Shandong First Medical University, Jinan, China; [2]Clinical Research and Development, Jiangsu Hengrui Pharmaceuticals Co., Ltd., Shanghai, China

山东第一医科大学附属中心医院
CENTRAL HOSPITAL AFFILIATED TO SHANDONG FIRST MEDICAL UNIVERSITY

Introduction

- Glucagon-like peptide 1 (GLP-1) receptor agonists are recognized for their ability to stimulate insulin secretion, suppress glucagon secretion, and delay gastric emptying.[1-2]
- Combining GLP-1 receptor agonists with other enteropancreatic hormones that exhibit complementary or synergistic actions, such as glucose-dependent insulinotropic polypeptide (GIP), a potent glucose-dependent stimulator of insulin secretion, may enhance metabolism more effectively.[4-5]
- Accumulating clinical evidence suggests that a dual GLP-1/GIP receptor agonist can achieve additive or synergetic effects on glycemic control and body weight loss by regulating both GLP-1 and GIP receptors.[6]
- HRS9531 is a novel agonist that exhibits remarkable activity on both GLP-1 and GIP receptors.
- We report the safety, tolerability, PK and PD of HRS9531 in healthy subjects.

Methods

- This is a phase 1, randomized, double-blind, placebo-controlled, SAD and MAD study (NCT05152277, **Figure 1**).
- In the SAD phase, healthy subjects were randomly assigned in a 4:1 ratio to receive subcutaneous injection of either HRS9531 (0.1, 0.3, 0.9, 2.7, 5.4, and 8.1 mg) or placebo.
- In the MAD phase, healthy subjects were randomly assigned in a 4:1 ratio to receive either HRS9531 (0.9, 2.7, and 5.4 mg [2.7/2.7/4.0/5.4 mg titration]) or placebo once a week for 4 weeks.
- The primary endpoints of both phases were safety and tolerability.

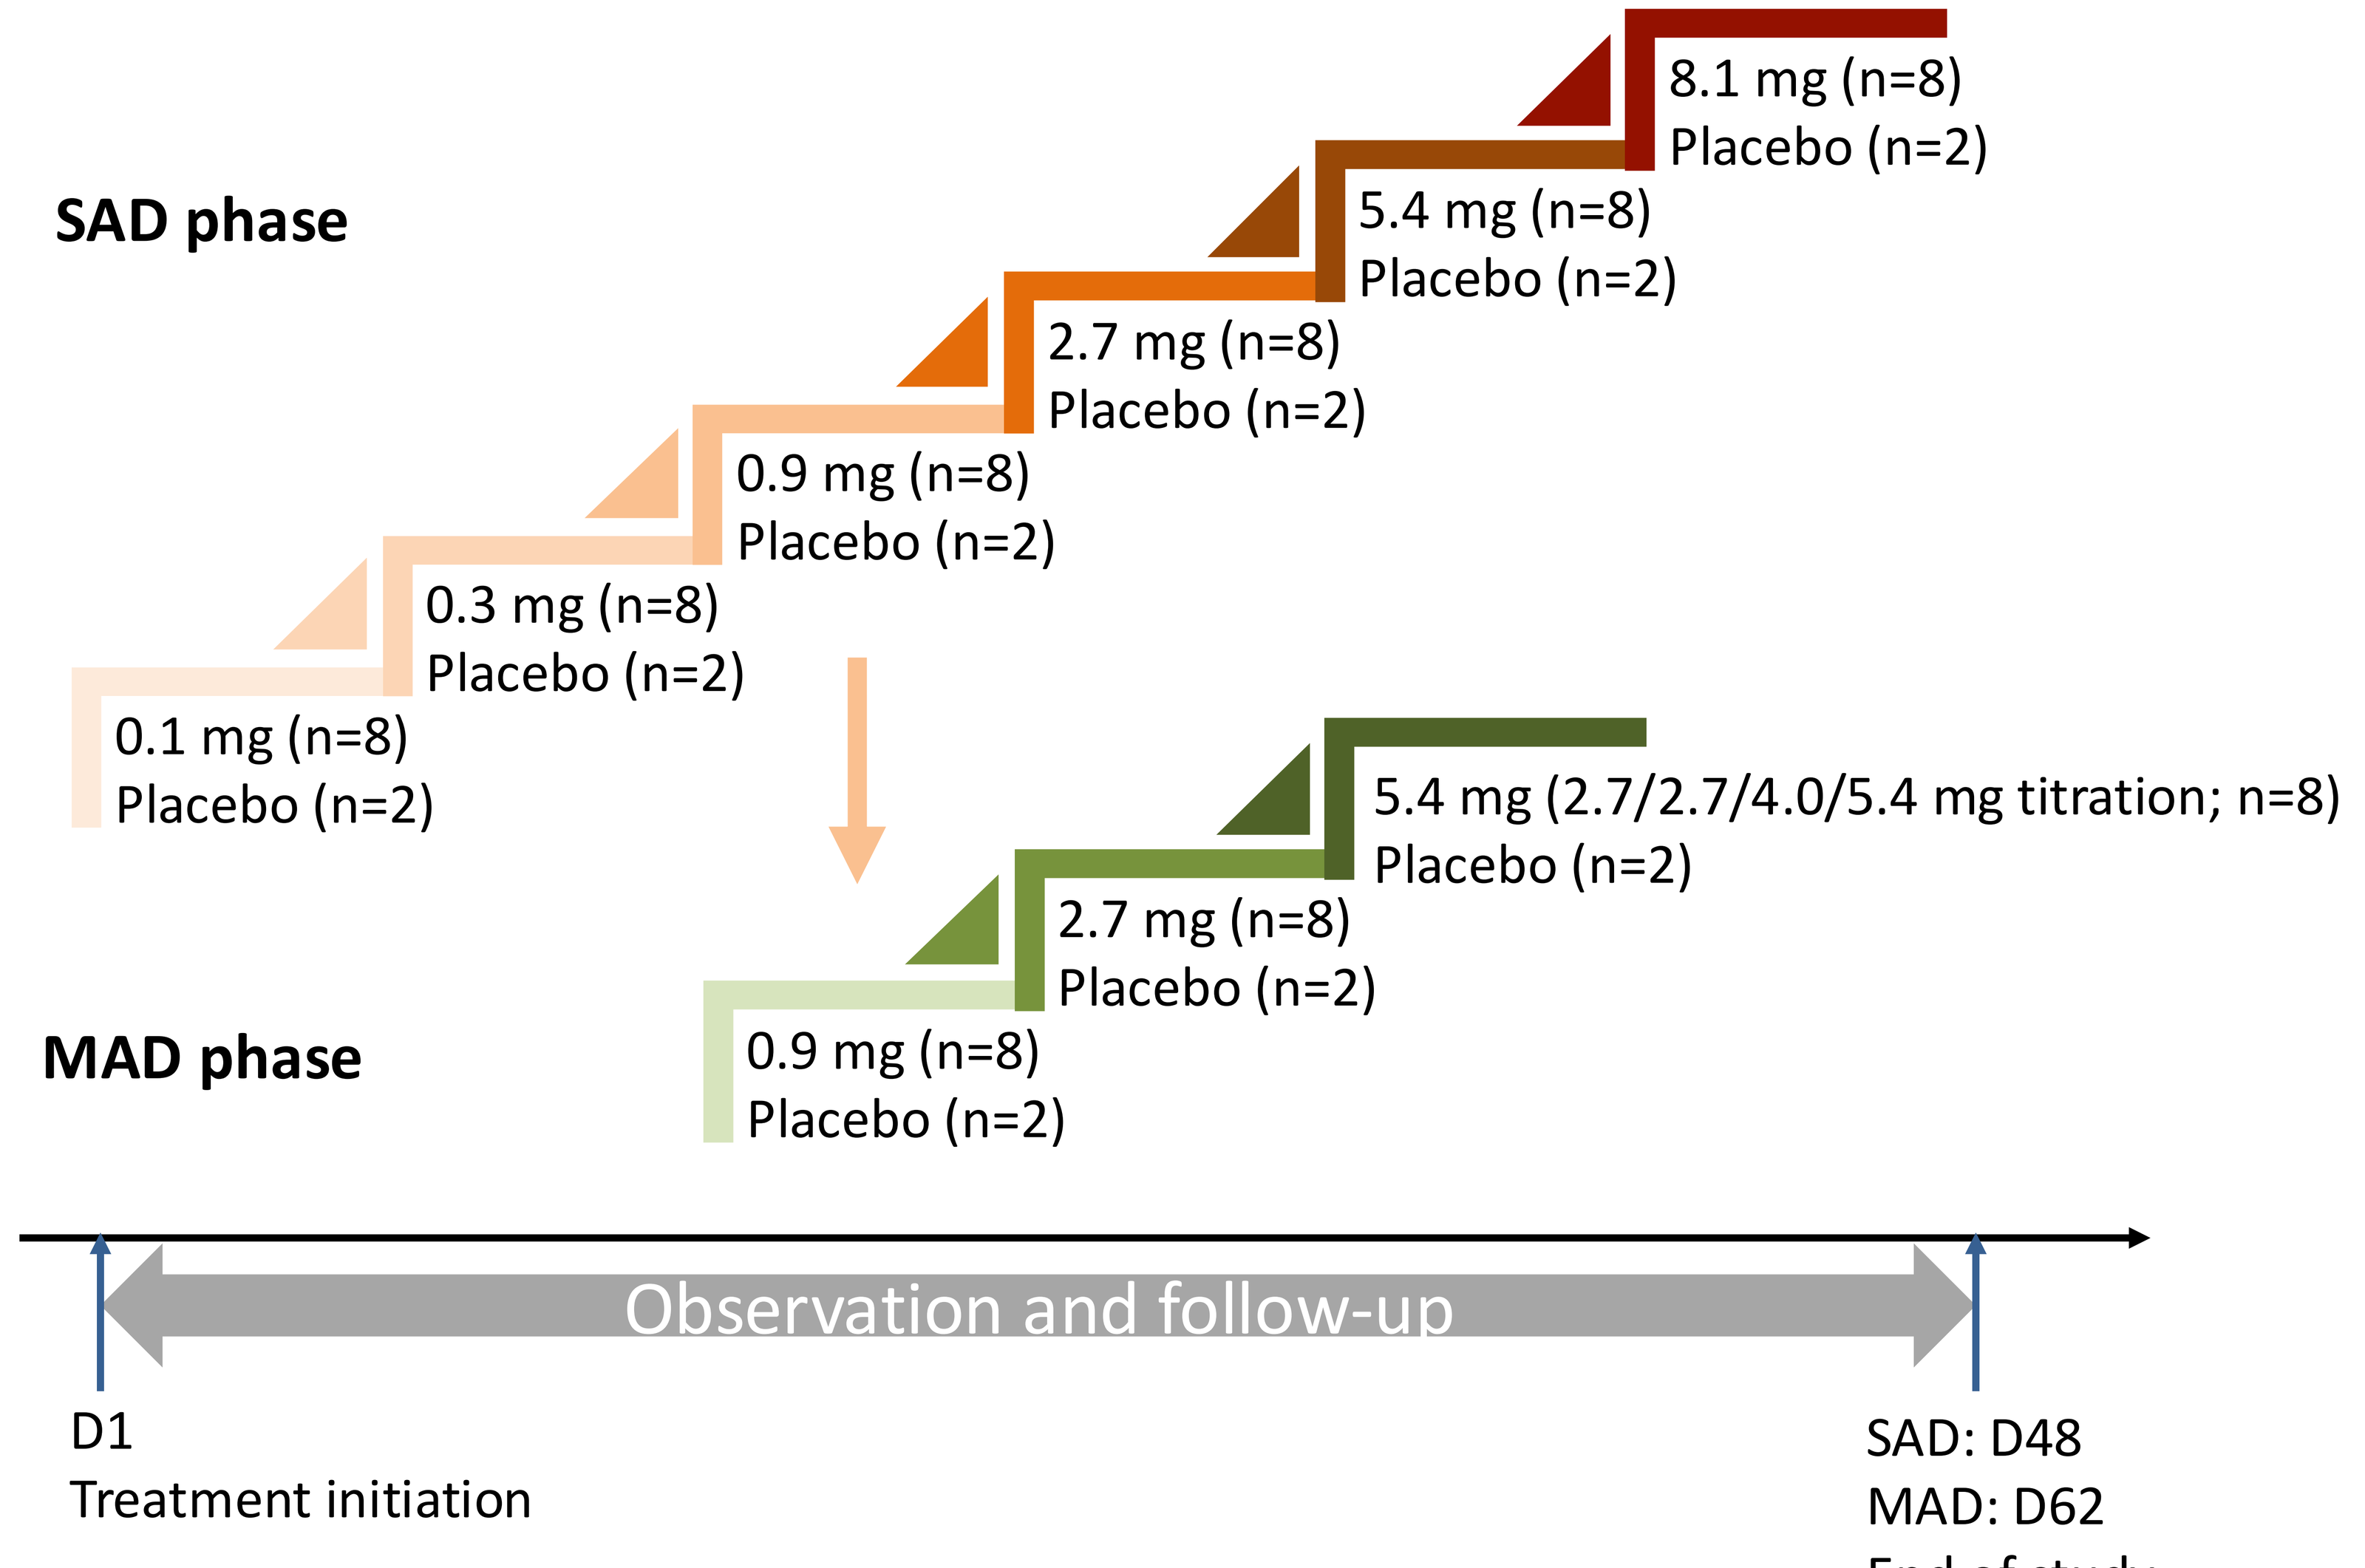
SAD phase
8.1 mg (n=8)
Placebo (n=2)
5.4 mg (n=8)
Placebo (n=2)
2.7 mg (n=8)
Placebo (n=2)
0.9 mg (n=8)
Placebo (n=2)
0.3 mg (n=8)
Placebo (n=2)
0.1 mg (n=8)
Placebo (n=2)
MAD phase
5.4 mg (2.7/2.7/4.0/5.4 mg titration; n=8)
Placebo (n=2)
2.7 mg (n=8)
Placebo (n=2)
0.9 mg (n=8)
Placebo (n=2)
Observation and follow-up
D1
Treatment initiation
SAD: D48
MAD: D62
End of study

Figure 1. Trial design

Results

Participants

- A total of 60 and 30 subjects were enrolled in the SAD and MAD phases, respectively (**Table 1**).

Table 1. Baseline characteristics

	SAD phase			MAD phase		
	Placebo (N=12)	HRS9531 (N=48)	SAD total (N=60)	Placebo (N=6)	HRS9531 (N=24)	MAD total (N=30)
Age, years	33.1 (9.0)	31.0 (9.6)	31.4 (9.5)	30.8 (8.6)	32.8 (8.3)	32.4 (8.3)
Male	7 (58.3)	34 (70.8)	41 (68.3)	6 (100)	17 (70.8)	23 (76.7)
Weight, kg	73.5 (8.8)	77.3 (12.6)	76.5 (11.9)	83.0 (5.6)	76.3 (14.3)	77.7 (13.2)
BMI, kg/m^2	27.1 (2.1)	27.2 (3.6)	27.2 (3.3)	27.8 (1.8)	26.8 (3.5)	27.0 (3.2)
HbA1c, %	5.5 (0.3)	5.5 (0.3)	5.5 (0.3)	5.5 (0.2)	5.4 (0.3)	5.4 (0.3)
FPG, mmol/L	4.9 (0.4)	5.0 (0.3)	5.0 (0.4)	5.0 (0.2)	4.9 (0.3)	4.9 (0.3)

Data are mean (SD) or n (%).

Safety

- The most common adverse events were abdominal distension and nausea in the SAD phase, and urine ketone body present and nausea in the MAD phase.
- All adverse events were mild to moderate in severity (mostly mild). No severe hypoglycemia or serious events were reported.

PK and PD

- Drug exposure after a single dose was approximately proportional to dose ranging from 0.9 to 8.1 mg (**Figure 2**).
- The median T_{max} and mean $t_{1/2}$ were 48–72 h and 156–182 h in the SAD phase, and 48–72 h and 169–192 h after the fourth dosing in the MAD phase, respectively.
- Fasting plasma glucose (FPG) levels decreased dose-dependently after both single and multiple dosing (**Figure 3**).
- The 0 to 2-hour glucose AUC during the oral glucose tolerance test (OGTT) on Day 23 decreased dose-dependently relative to the placebo in the MAD phase (**Figure 4**).
- In the SAD phase, dose-dependent body weight loss was observed, with the maximum mean loss (3.8 kg, 4.9%) occurring in the 8.1 mg group on Day 8. In the MAD phase, the mean weight loss on Day 29 (after 4 weeks of treatment) ranged from 4.3–7.7 kg (6.7%–9.3%) across the 0.9–5.4 mg groups, with the maximal mean loss (8.0 kg, 10.0%) occurring in the 5.4 mg group on Day 36 (**Figure 5**).

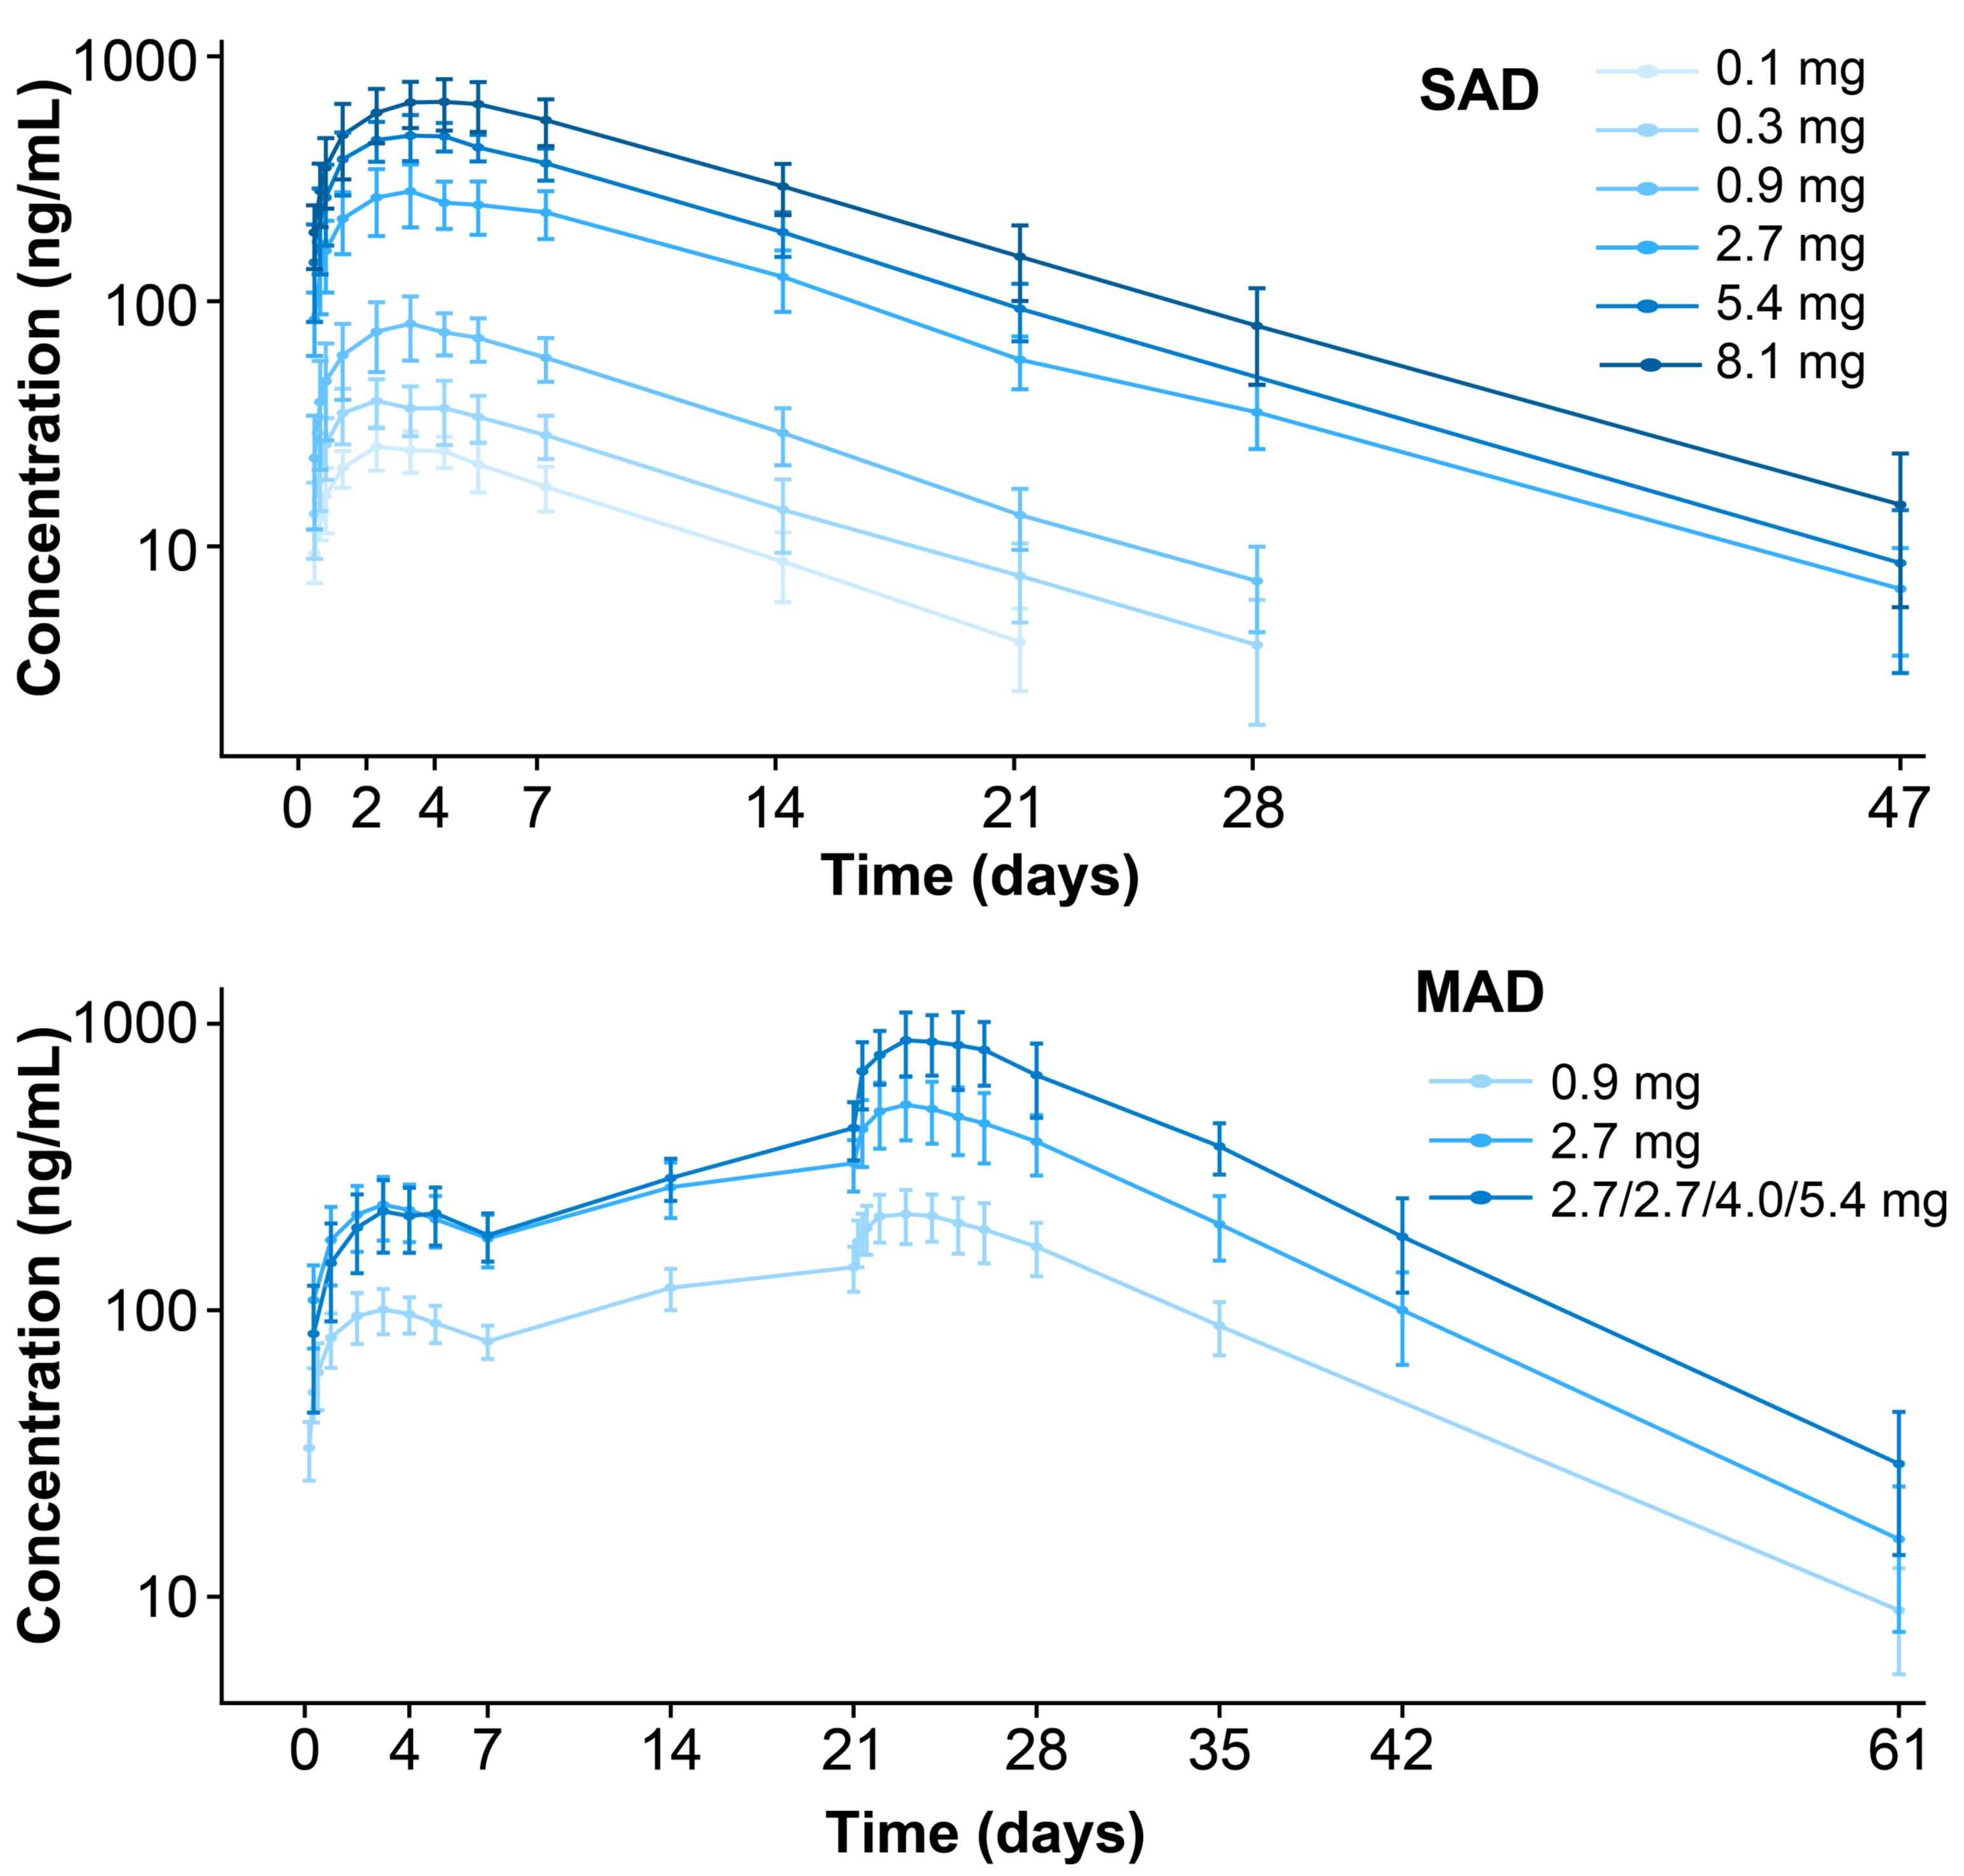
SAD
0.1 mg
0.3 mg
0.9 mg
2.7 mg
5.4 mg
8.1 mg
Concentration (ng/mL)
1000
100
10
0 2 4 7 14 21 28 47
Time (days)
MAD
0.9 mg
2.7 mg
2.7/2.7/4.0/5.4 mg
Concentration (ng/mL)
1000
100
10
0 4 7 14 21 28 35 42 61
Time (days)

Figure 2. Mean PK concentration-time curve

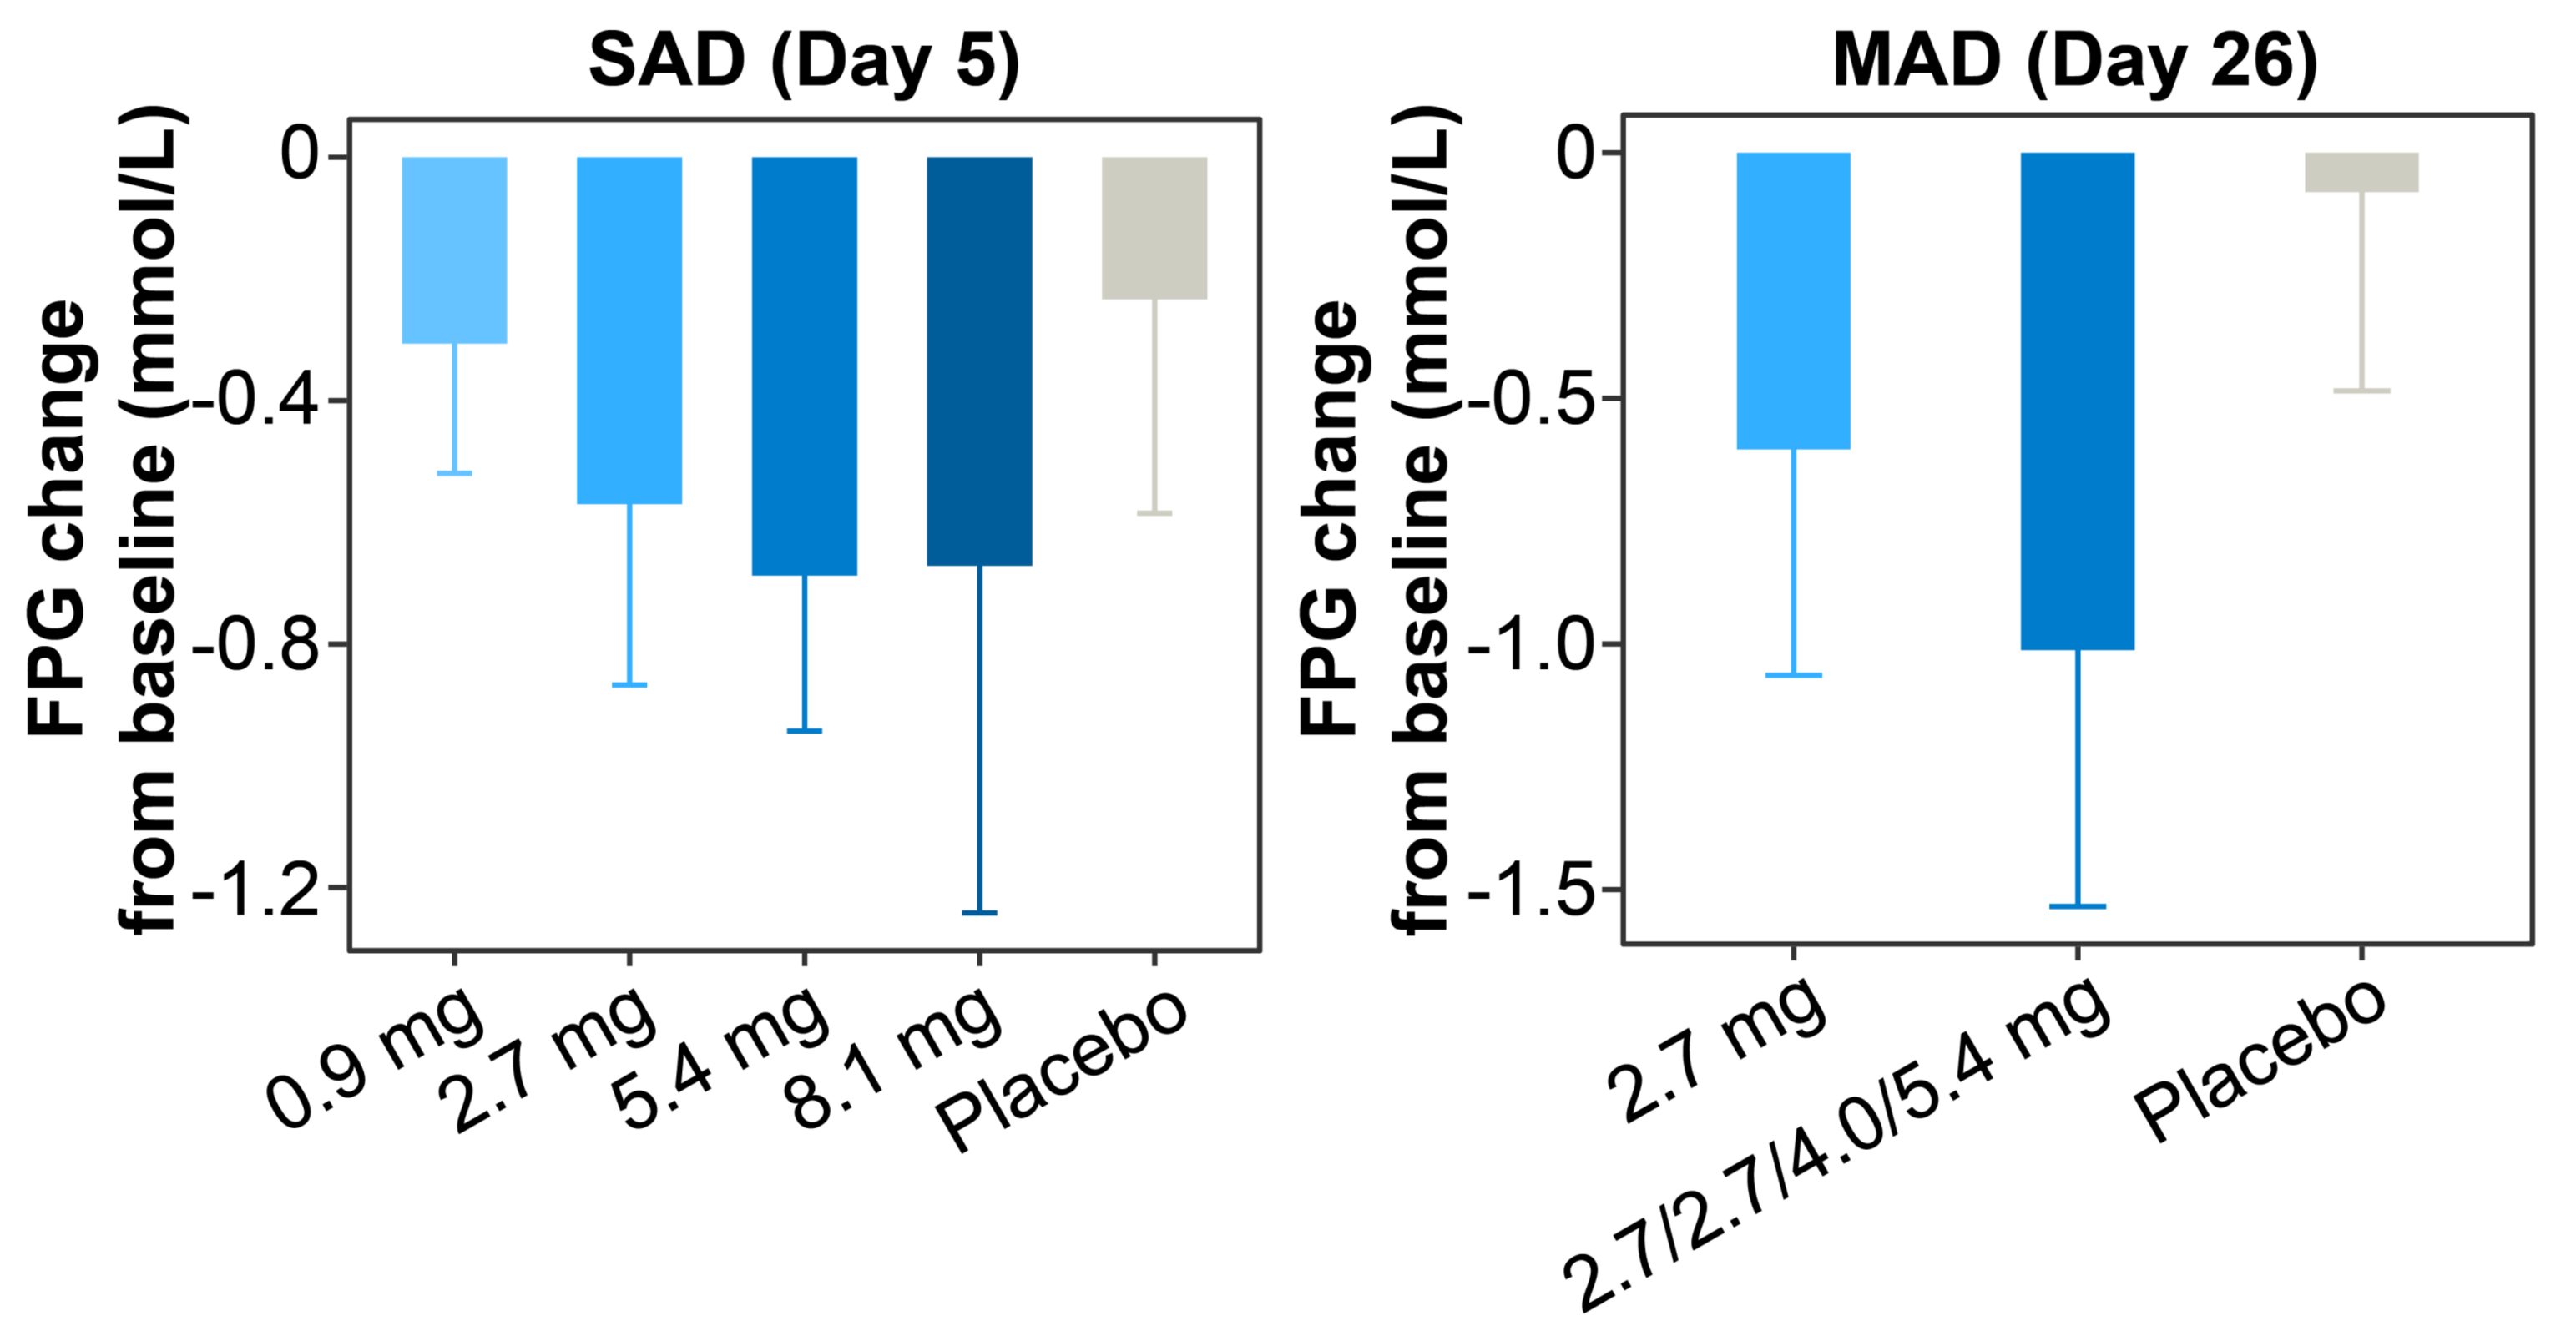
SAD (Day 5)
FPG change from baseline (mmol/L)
0
-0.4
-0.8
-1.2
0.9 mg
2.7 mg
5.4 mg
8.1 mg
Placebo
MAD (Day 26)
FPG change from baseline (mmol/L)
0
-0.5
-1.0
-1.5
2.7 mg
2.7/2.7/4.0/5.4 mg
Placebo

Figure 3. Mean FPG concentration change from baseline

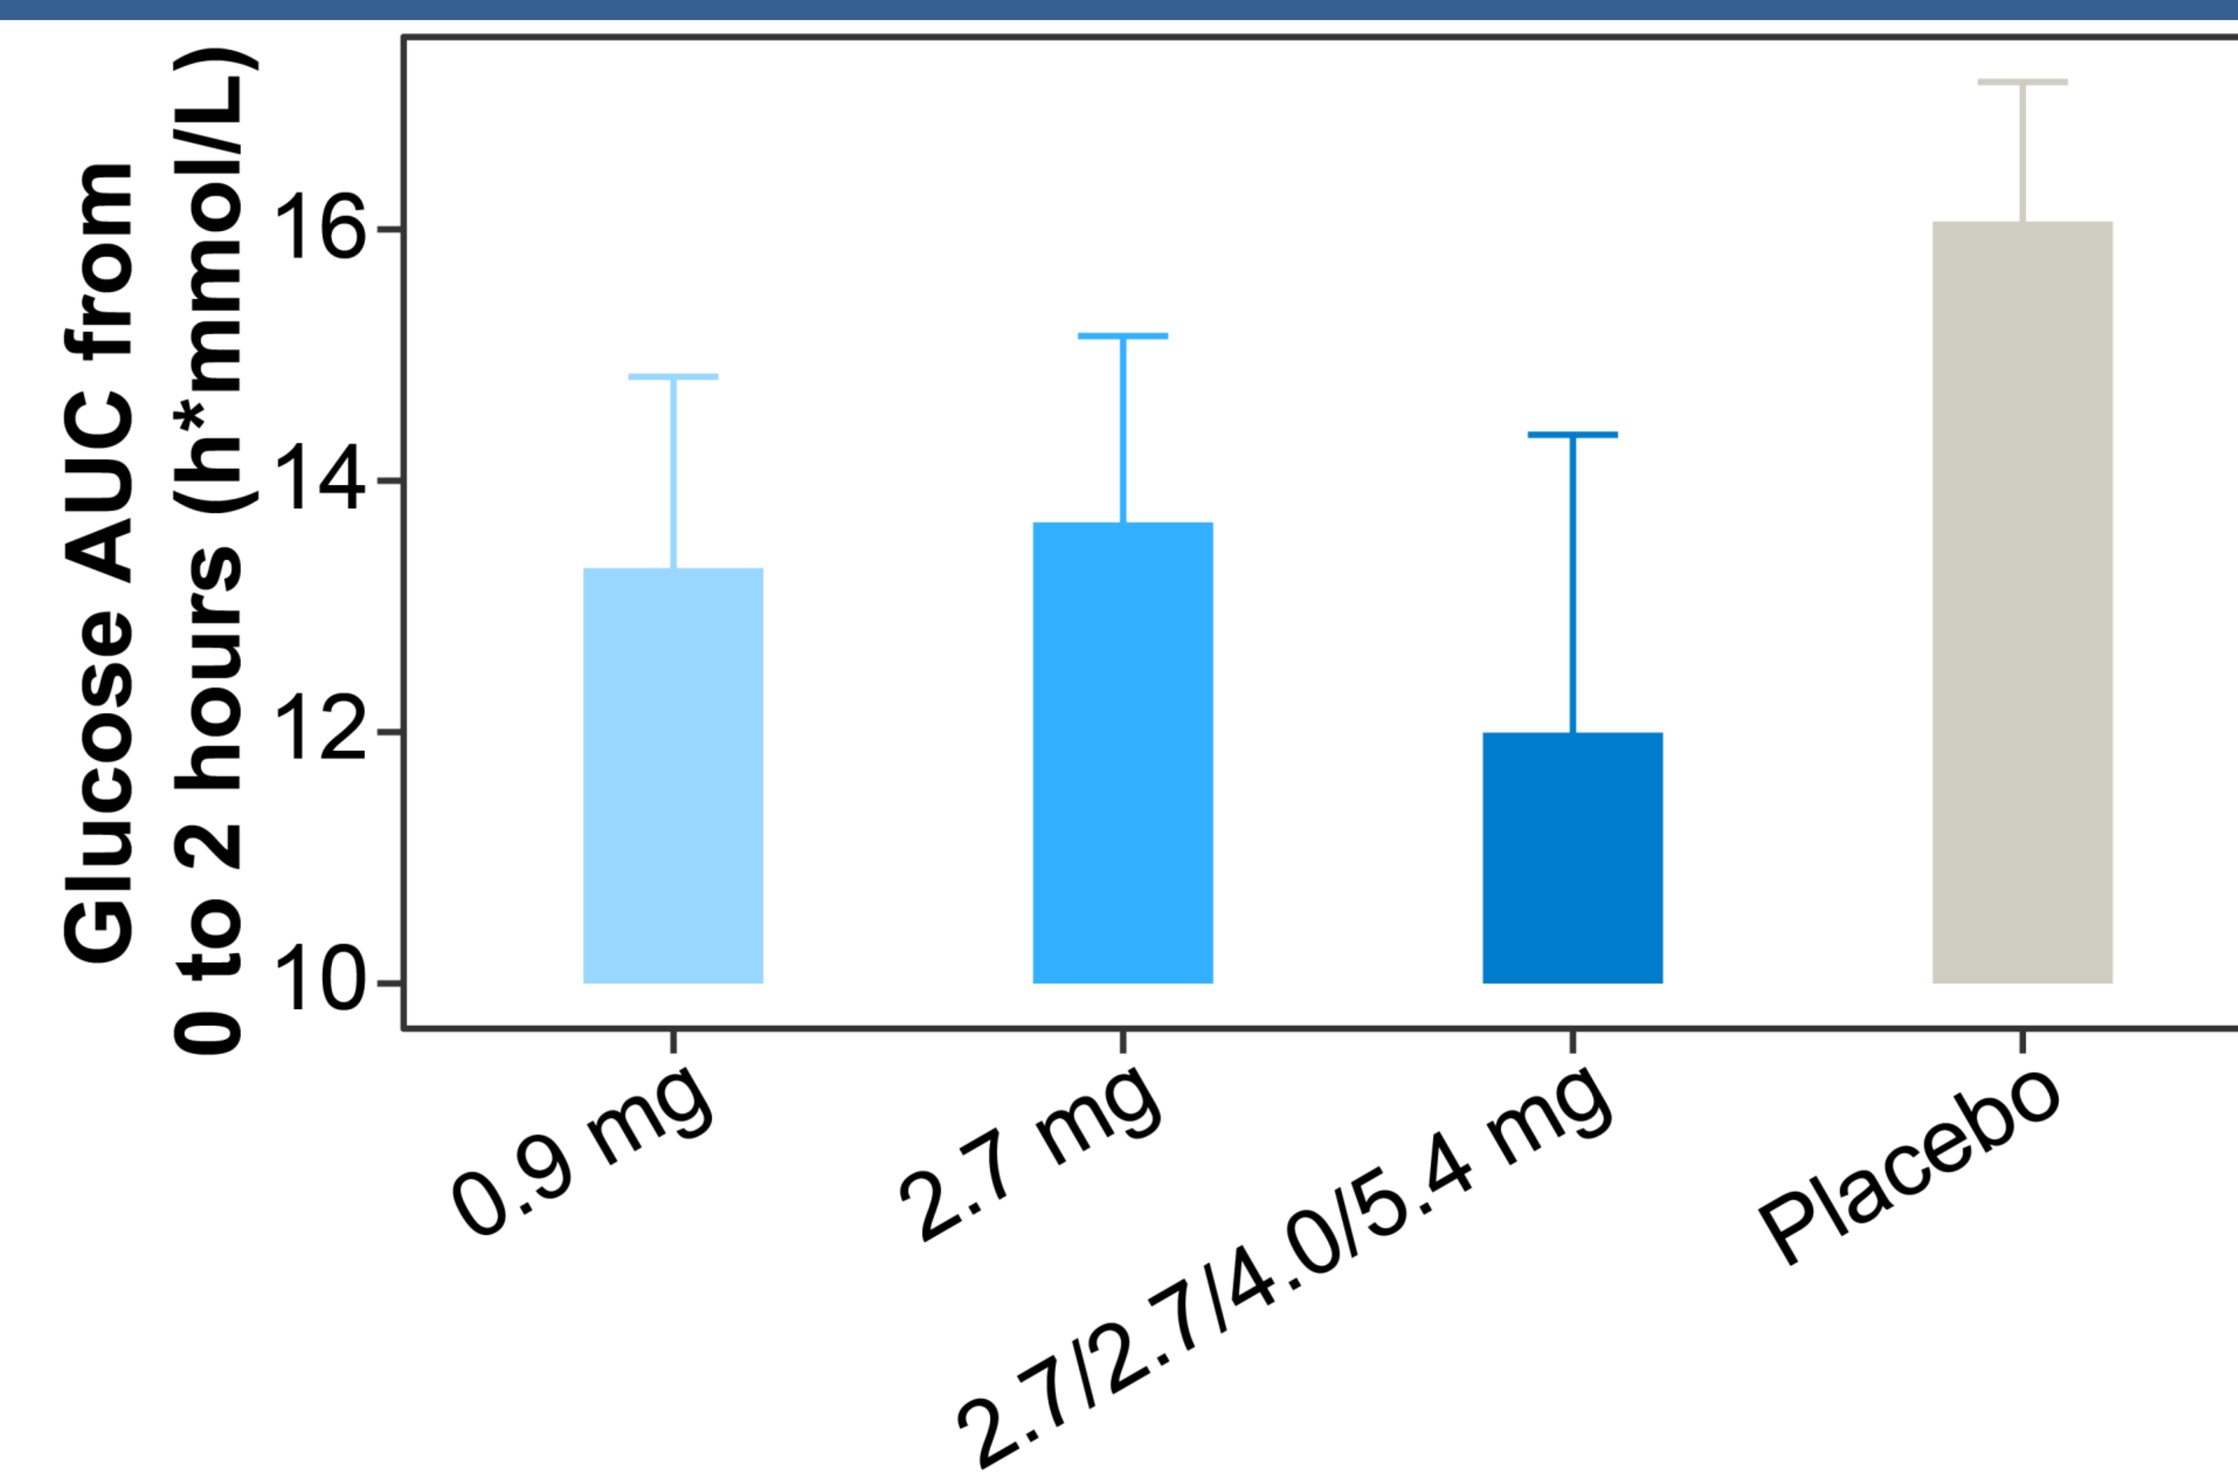
Glucose AUC from 0 to 2 hours (h*mmol/L)
16
14
12
10
0.9 mg
2.7 mg
2.7/2.7/4.0/5.4 mg
Placebo

Figure 4. Mean glucose concentration during OGTT

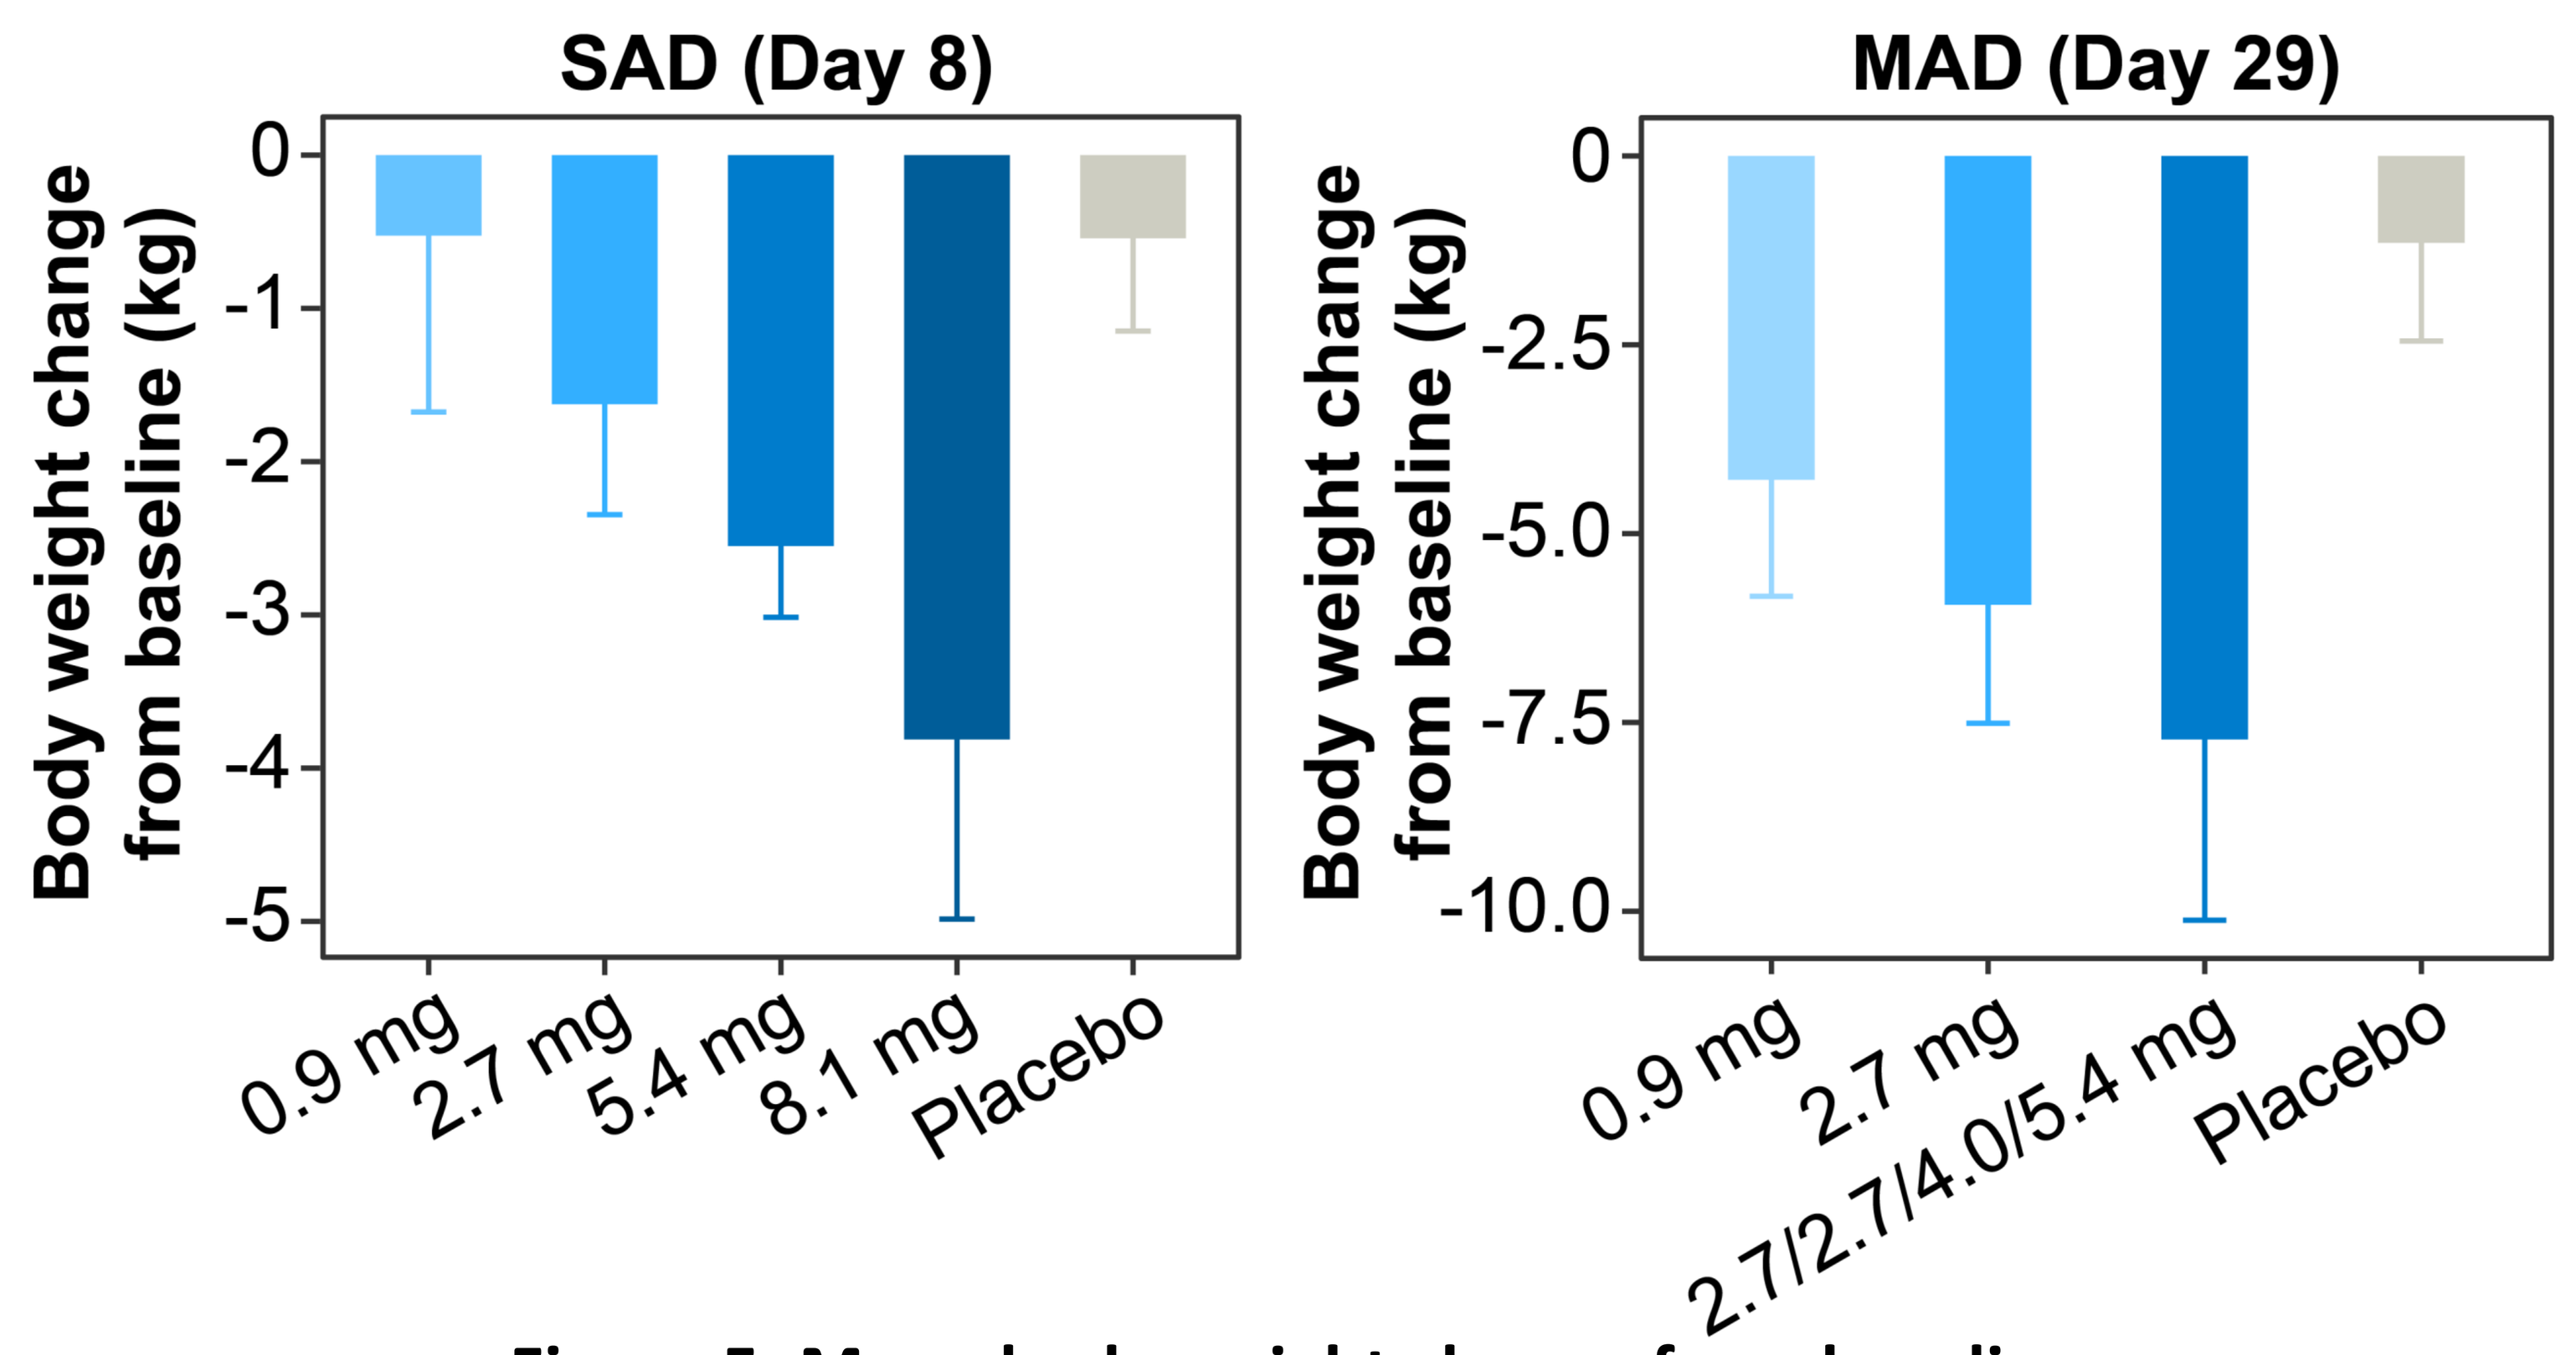
SAD (Day 8)
Body weight change from baseline (kg)
0
-1
-2
-3
-4
-5
0.9 mg
2.7 mg
5.4 mg
8.1 mg
Placebo
MAD (Day 29)
Body weight change from baseline (kg)
0
-2.5
-5.0
-7.5
-10.0
0.9 mg
2.7 mg
2.7/2.7/4.0/5.4 mg
Placebo

Figure 5. Mean body weight change from baseline

Conclusions

- HRS9531 was well-tolerated, exhibited linear PK, and possessed a long elimination half-life. Additionally, it led to evident reductions in glucose levels and body weight compared to placebo.
- These data supported the further clinical development of HRS9531 as a potential therapeutic option for metabolism-related disorders such as T2DM and obesity.

Conflicts of Interest

- Qing Wen has nothing to declare.

Acknowledgements

- The patients and their families.
- The investigators and clinical study sites.
- The study is sponsored by Jiangsu Hengrui Pharmaceuticals Co., Ltd.

References: 1. Drucker, et al. Lancet 2006; 368: 1696–705; 2. Nauck, et al. Lancet Diabetes Endocrinol 2016; 4: 525–36; 3. Holst, et al. Physiol Rev 2007; 87: 1409–39; 4. Marso, et al. N Engl J Med 2016; 375: 311–22. 5. Tuttle, et al. Lancet Diabetes Endocrinol 2018; 6: 605–17; 6. Rosenstock, et al. Lancet 2021; 398: 143–55.

Contact: Qing Wen, MD (wenq0619@126.com)